Frequency-domain derived optimisation of cell-cycle specific cancer treatment

Pascal Schulthess[1], James Yates[2], and Piet Hein van der Graaf[1,3]

Systems Biomedicine & Pharmacology

INTRODUCTION

It was recently shown that key biological control systems (e.g. MAPK pathway) are highly sensitive to the frequency of external stimuli in a non-intuitive manner which cannot be predicted by conventional pharmacometrics approaches [1]. This suggests that quantitative systems pharmacology (QSP) can provide novel insights into optimal dosing regimens which could add a new dimension to the design of novel treatments. However, methods for such an approach are currently lacking. Recently, we illustrated the utility of frequency-domain response analysis (FdRA), a method widely used engineering, using several generic PK/PD case studies [2]. We now demonstrate the use of FdRA to optimise treatment regimen for cell-cycle specific chemotherapy.

CONCLUSION

Frequency-domain response analysis...

... identifies drug dosing frequencies for which the plasma concentration amplitude is attenuated/amplified in the response.

... identifies effective and save drug dosing frequencies for the cell-cycle specific tumour growth model.

... **suggest metronomic chemotherapy as optimal cancer treatment.**

METHODS

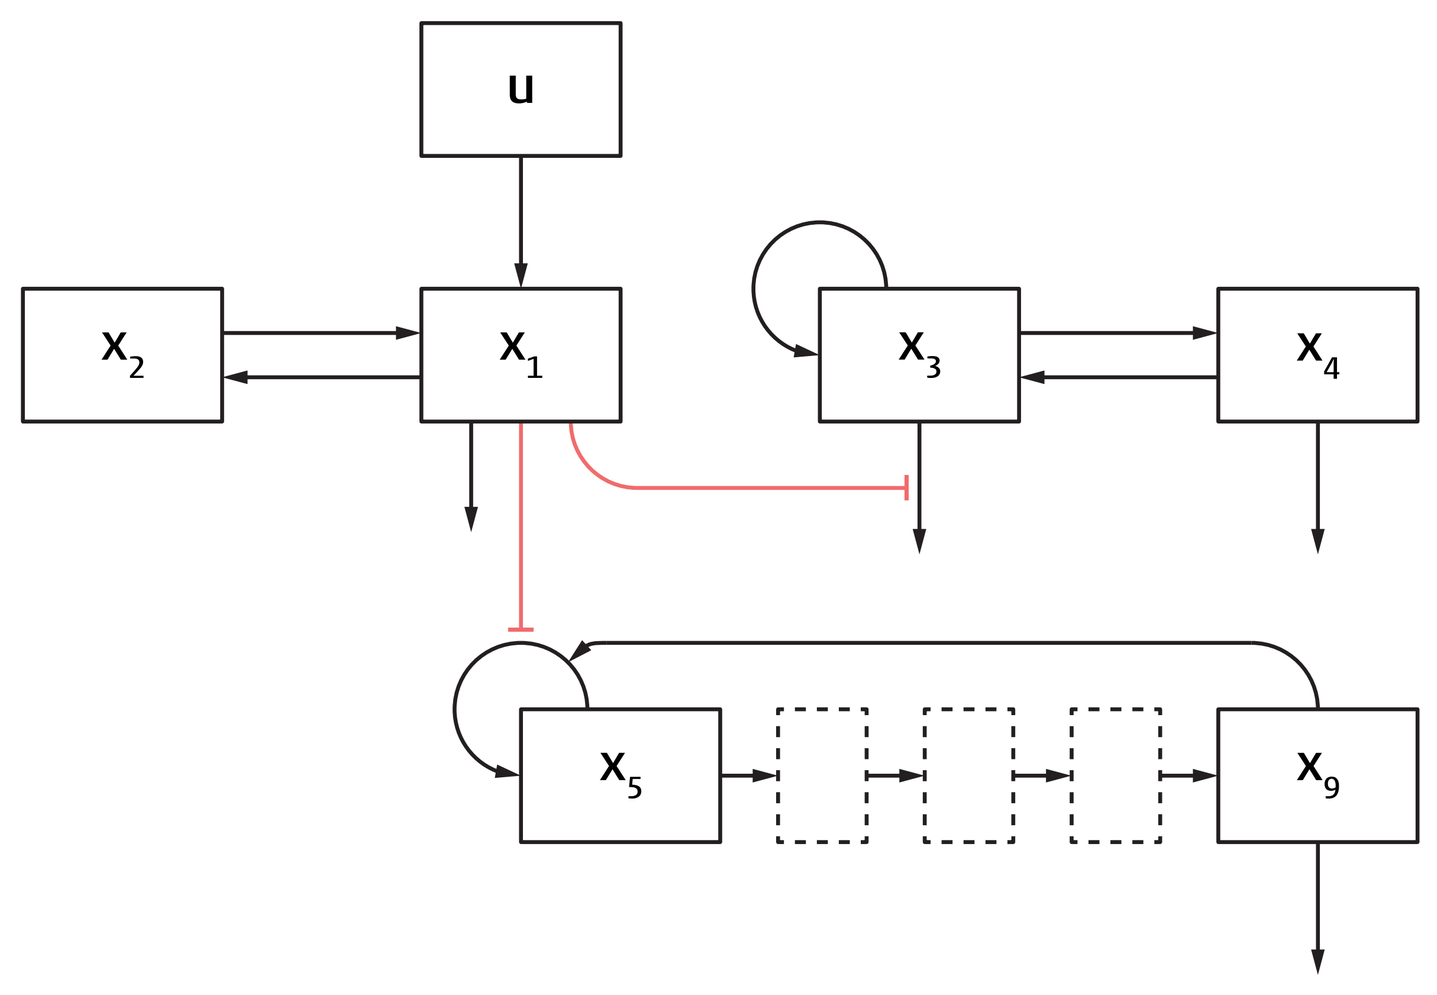
u
x_2
x_1
x_3
x_4
x_5
x_9

Figure 1: Two-comp. PK of Etoposide (x_1, x_2) linked to cell-cycle specific cancer model (x_3, x_4) and Fribergs model of myelosuppression (x_5 to x_9).

Analytically, frequency-domain response analysis (FdRA) determines the input/output behaviour of a linear, time-invariant (LTI) system

$$\dot{\boldsymbol{x}} = \boldsymbol{A}\boldsymbol{x} + \boldsymbol{b}u$$
$$y = \boldsymbol{c}^T\boldsymbol{x} + du$$

in response to sinusoidal inputs of varying frequency. Thus, for the LTI system with $\boldsymbol{x}$, y, and u denoting the model states, the outputs, and the inputs, respectively, and with $\boldsymbol{x}(0) = \boldsymbol{x}_0$ a so-called transfer funtion can be defined as

$$G(s) = \frac{Y(s)}{U(s)} = \boldsymbol{c}^T\left(s\boldsymbol{I} - \boldsymbol{A}\right)^{-1}\boldsymbol{b} + d$$

The transfer function can now be used to collect the responses of the LTI system to sinusoidal inputs over a wide range of frequencies $s = i\omega$ with the help of a Bode plot . Its magnitude is defined as:

$$M(\omega) = \log_{10}|G(i\omega)|$$

and represents the logarithmic ratio of the output and the input amplitude.

Numerically, the frequency response can be determined for non-sinusoidal inputs that arise from repetitive dosing regimen commonly used in QSP modelling [2]. We here apply FdRA to a PK-linked cell-cycle specific tumour growth model while considering safety by a model of myelosuppression (Figure 1).

The two-compartment PK model of Etoposide [3] is given by:

$$\dot{x}_1 = k_{21}x_2 - (k_{12} + k_{\mathrm{e}})x_1$$
$$\dot{x}_2 = k_{12}x_1 - k_{21}x_2$$

wherein x_1 and x_2 describe the plasma and peripheral compartment, respectively. Tumour growth dynamics are modeled by a cell-cycle specific two compartment model [4] in which proliferating cancer cells in G_1, S, G_2, and M phase of the cell cycle are given by x_3 while quiescent cancer cells in G_0 phase are denoted by x_4

$$\dot{x}_3 = (\alpha - \mu - \eta)x_3 + \beta x_4 - \frac{k_1}{V_1}x_1x_3$$
$$\dot{x}_4 = \mu x_3 - (\beta + \gamma)x_4$$

Fribergs model of myelosuppression [5] describes stem and progenitor cells (x_5), three transit compartments of maturing cells (x_6, x_7, and x_8), and circulating neutrophils (x_9) by:

$$\dot{x}_5 = k_\tau x_5\left(\delta(x_1)\frac{x_{90}^\lambda}{x_9^\lambda} - 1\right)$$
$$\dot{x}_6 = k_\tau(x_5 - x_6)$$
$$\dot{x}_7 = k_\tau(x_6 - x_7)$$
$$\dot{x}_8 = k_\tau(x_7 - x_8)$$
$$\dot{x}_9 = k_\tau(x_8 - x_9)$$

RESULTS

The cell-cycle specific tumour growth model is now stimulated accordint to different dosing schemes. Examplified in Figure 2, we assume constant exposure to Etoposide at the maximum tolerated dose of 500 mg/ m^2 either at a dosing frequency of one dose every 60 days (blue lines) or at a dosing frequency of one dose every 7 days (green lines).

We observe that even though higher doses are administered every 60 days the tumour continues to grow while low doses every 7 days lead to overall decrease in tumour mass. Additionally, more frequent lower doses do not disrupt neutrophil count as strongly as less frequent higher doses.

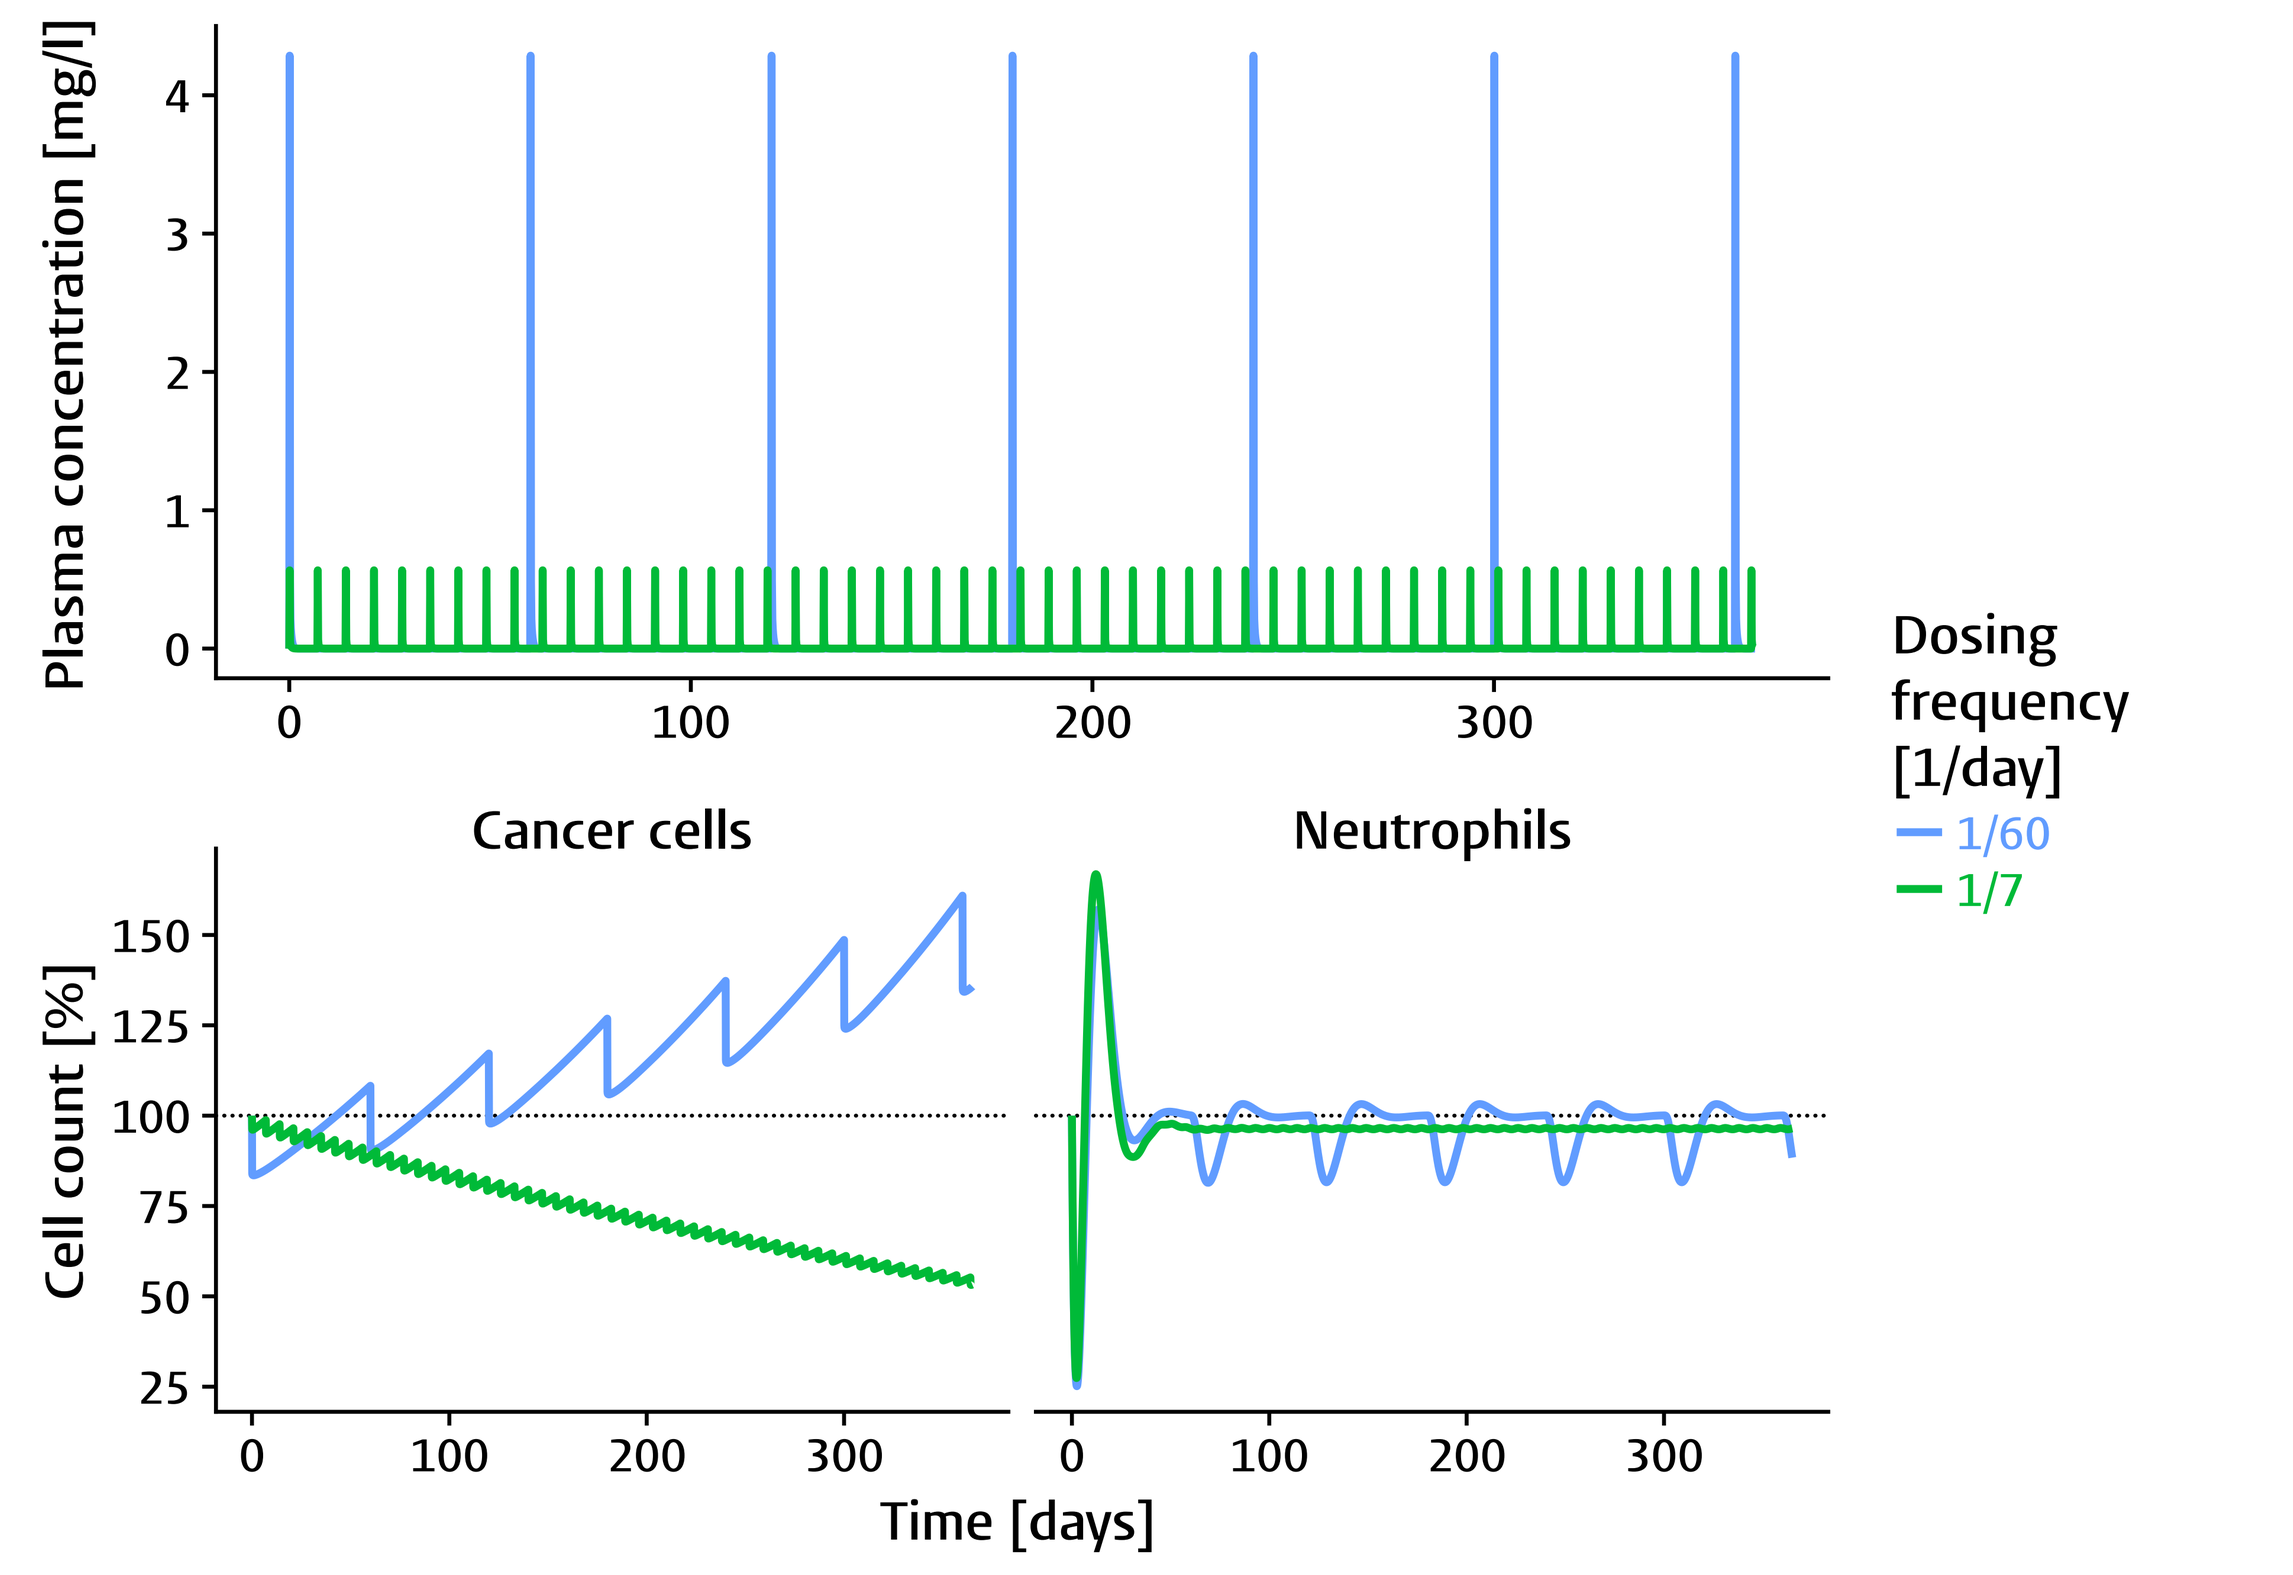
Plasma concentration [mg/l]
0
1
2
3
4
0
100
200
300
Dosing frequency [1/day]
1/60
1/7
Cancer cells
Neutrophils
Cell count [%]
25
50
75
100
125
150
Time [days]

Figure 2: Cancer cell and neutrophil time courses for two dosing frequencies at constant exposure showing effective (green) and ineffective (blue) dosing.

In order to systematically study the response of cancer cells and neutrophils to different dosing schemes, we applied numerical FdRA to the model given in Figure 1. To this end, we administered doses at frequencies between one bolus dose every 120 days and 24 bolus doses per day at either variable (i.e. each dose equals 500 mg/ m^2) or constant exposure. Afterwards, we measured the amplitude of the plasma concentration of Etoposide as well as the amplitudes of the total number of cancer cells (proliferating + quiescent cells) and neutrophils after one year of treatment. The output to input amplitude ratio is then plotted over the dosing frequency in Figure 3.

We observe that only constant exposure negatively affects safety (i.e. neutrophil counts below $0.5{\cdot}10^9$). Furthermore, less frequent dosing is ineffective and typically leads to a higher amplitude ratio. Only the neutrophil amplitude ratio under variable exposure increases for decreasing dosing frequencies while in all other cases the amplitude ratio decreases.

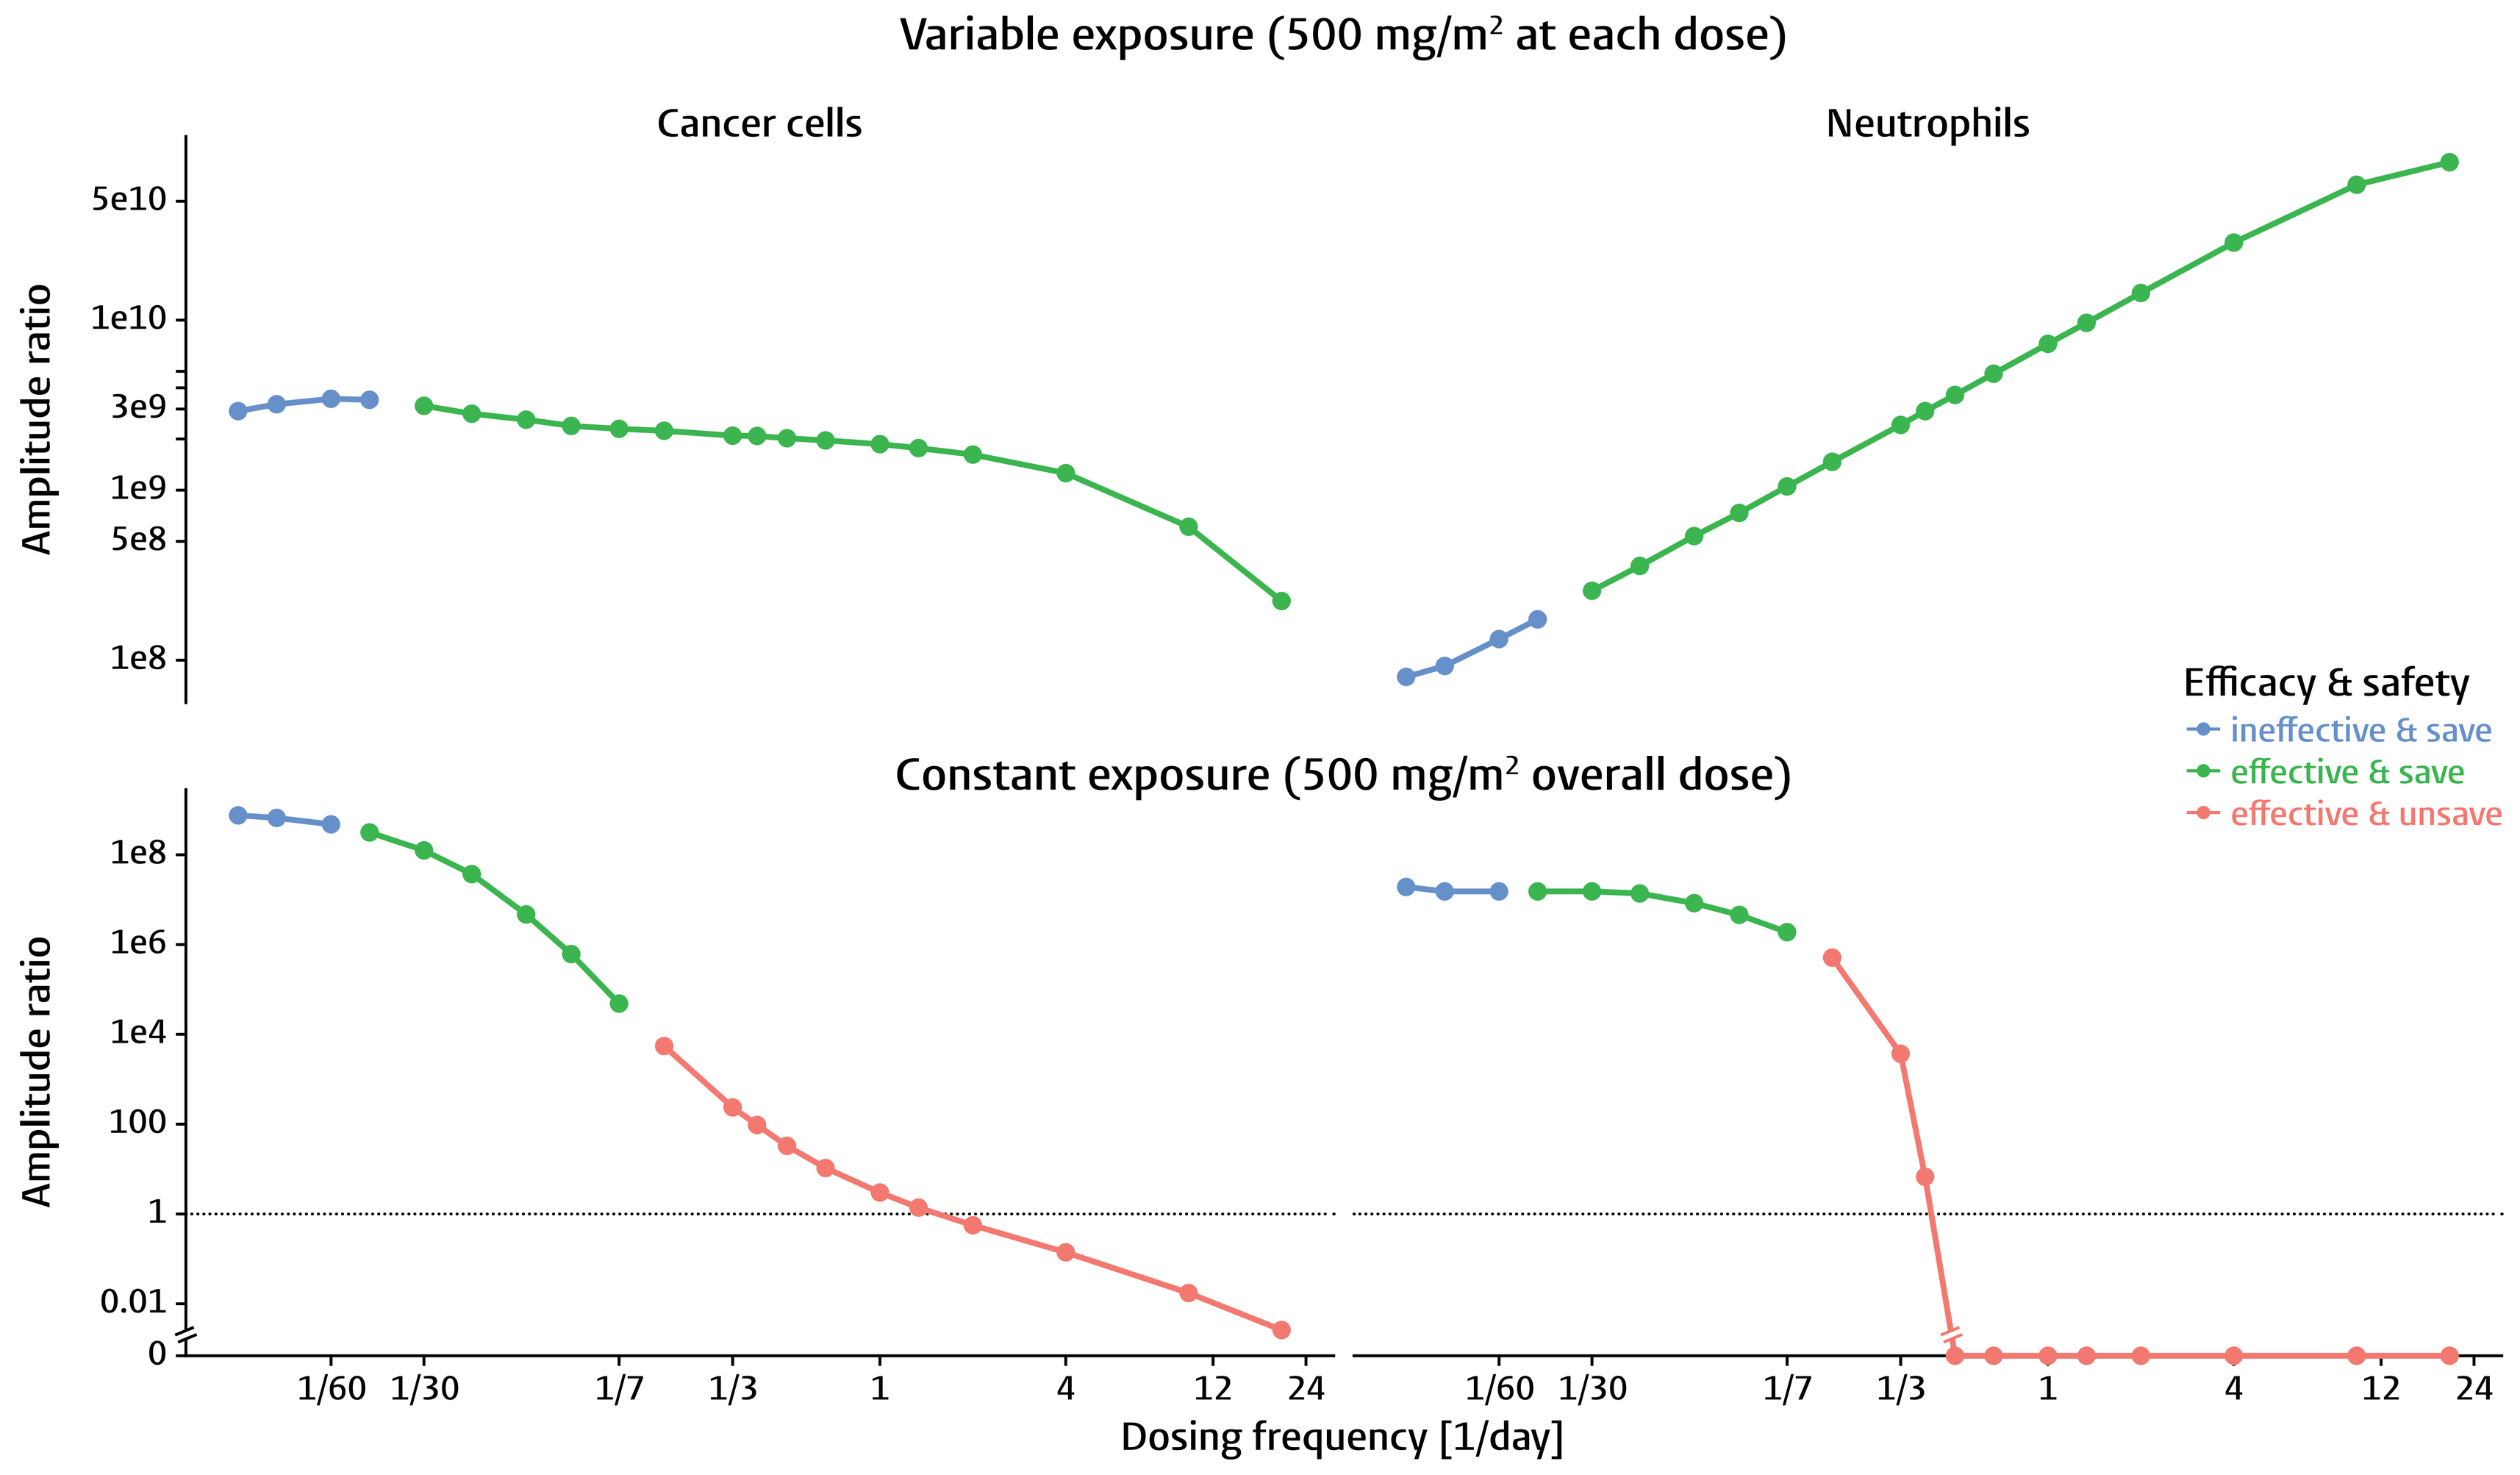
Variable exposure (500 mg/m² at each dose)
Cancer cells
Neutrophils
Amplitude ratio
5e10
1e10
3e9
1e9
5e8
1e8
Efficacy & safety
ineffective & save
effective & save
effective & unsave
Constant exposure (500 mg/m² overall dose)
Amplitude ratio
1e8
1e6
1e4
100
1
0.01
0
1/60
1/30
1/7
1/3
1
4
12
24
Dosing frequency [1/day]

Figure 3: Cancer cell and neutrophil frequency response for constant and variable exposure.

AFFILIATIONS & FUNDING

1. Systems Biomedicine & Pharmacology, LACDR, Leiden University, Leiden, The Netherlands
2. DMPK, Oncology, Innovative Medicines and Early Development, AstraZeneca, Chesterford Research Park, United Kingdom
3. Certara QSP, Canterbury Innovation House, Canterbury, United Kingdom

REFERENCES

[1] Mitchell, A., Wei, P. & Lim, W. A. Science 350, 1379–1383 (2015).
[2] Schulthess, P. et al. CPT Pharmacometrics Syst Pharmacol 7, 111–123 (2017).
[3] Zhu, J. et al. J Biomed Inform 57, 20–27 (2015).
[4] Panetta, J. C. & Adam, J. Mathematical and Computer Modelling 22, 67–82 (1995).
[5] Friberg, L. E. et al. Journal of Clinical Oncology 20, 4713–4721 (2002).

Discover the World at Leiden University